Efficacy of the mouthwash CB12 in the treatment of intra-oral halitosis: a double-blind, randomised, controlled trial

Rainer Seemann[1], Andreas Filippi[2], Sebastian Michaelis[3], Susanne Lauterbach[4], Hans-Dieter John[5], Jörg Huismann[6]
[1]Department of Preventive-, Restorative- and Pediatric Dentistry, zmk bern, University Bern, Switzerland; [2]Klinik für Zahnärztliche Chirurgie, -Radiologie, Mund- und Kieferheilkunde und Zahnunfallzentrum, Universitätskliniken für Zahnmedizin, Hebelstr. 3, CH-4056 Basel; [3]Zahnarztpraxis Nordstrasse, Düsseldorf, Germany; [4]Zahnfitness Lauterbach, St Sebastian, Germany; [5]Periodontics Düsseldorf, Düsseldorf, Germany; [6]Köln-Deutz, Germany

Introduction

- The prevalence of halitosis in the general public is estimated at around 30%.(1-4) Around 90% of these halitosis cases are caused by volatile sulphur compounds (VSC) produced by anaerobic bacteria in the oral cavity (intra oral halitosis).(5;6)
- Organoleptic scoring by a trained odour judge is considered the gold standard for assessing breath odour,(7) but is subjective and difficult to standardise.(6) The OralChroma gas chromatograph and the Halimeter measure VSCs objectively and correlate well with organoleptic assessment.(8;9)
- CB12 (MEDA OTC, Sweden) is an over-the-counter mouthwash containing zinc acetate (0.3%) and low concentration chlorhexidine (0.025%).
- The efficacy of CB12 in treating halitosis has been shown in several open label studies:
 - CB12 reduced halitosis more effectively than placebo (i.e. significantly greater inhibition of VSC production), without causing tooth discolouration, mucosal lesions or taste disturbance.(10)
 - CB12 also reduced halitosis more effectively than five other mouthwash products (assessed by both organoleptic score and inhibition of VSC production) with superiority noted for up to 3 hours.(11)

Aim

- To establish the long-term (12 hour) efficacy of CB12 in a rigours state-of-the-art randomised, placebo-controlled, double blind, cross-over study in subjects with intra-oral halitosis.

Methods

- Eligible subjects were otherwise healthy but had halitosis of intra-oral origin with daily periods of noticeable halitosis; had an organoleptic score ≥2 and a total VSC >160 ppb, hydrogen sulphide ≥112 ppb and methyl mercaptan ≥26 ppb prior to first dose.
- Exclusion criteria included periodontitis, open caries lesions, obvious gingival inflammation or gingivitis, and oral thrush.

Subjects

- Eligible subjects were otherwise healthy but had halitosis of intra-oral origin with daily periods of noticeable halitosis; had an organoleptic score ≥2 and a total VSC >160 ppb, hydrogen sulphide ≥112 ppb and methyl mercaptan ≥26 ppb prior to first dose.
- Exclusion criteria included periodontitis, open caries lesions, obvious gingival inflammation or gingivitis, and oral thrush.

Study design

- Randomised, controlled, double-blind, two-sequence, three-fold replicate, crossover design
- Subjects were randomised 1:1 to receive:
 - CB12-water-water OR
 - water-CB12-CB12
- The second period was replicated to minimise possible carry-over effects between periods
- Treatments were taken 12 hours apart and there was a 5-day washout between periods.
- The study was conducted in agreement with the ICH guideline on Good Clinical Practice (ICH E6) and other applicable guidelines.

Interventions

- All treatments were administered by rinsing 10 ml in the mouth for 30 seconds, as per the user instructions for CB12. The control was non-carbonated water.
- Treatments were administered under supervised, controlled conditions.
 - Subjects stayed in the study ward for the observational period of about 30 hours
 - Smoking, foods associated with oral malodour, antibacterial lozenges and changes in oral hygiene practices were not permitted on study days or the preceding 48 hours.

Efficacy assessment

- Assessments were made at
 - Baseline (i.e. in the evening prior to first mouth rinse),
 - 12 hours after first treatment (i.e. in the morning following night sleep; overnight assessment), and
 - 12 hours after second treatment (i.e. in the evening 24 hours after the first treatment; day assessment).
- Oral levels of hydrogen sulphide, methyl mercaptan and dimethyl sulphide were measured by OralChroma.
- Total oral VSC concentration was assessed by Halimeter.
- Organoleptic score was assessed by a trained odour judge using the 0-5 scale by Rosenberg (modified by Greenman): 0 = no odour; 1 = barely noticeable odour; 2 = slight odour; 3 = moderate odour; 4 = strong odour; 5 = very strong odour (saturation).(12)

Statistical analyses

- The primary efficacy measure was hydrogen sulphide concentration and the co-primary measure was methyl mercaptan concentration, both after 12 hours (overnight assessment).
- The key secondary outcome measure was the change in organoleptic score from 0 hours to 12 hours.
- Analysis was performed for the intention-to-treat population (ITT; all subjects randomised and exposed to study treatment who had at least one assessment of efficacy), with a one-sided α-level of 2.5%.
- Safety was analysed in all randomised subjects who received at least one dose of study treatment.

Results

Baseline characteristics

- 34 subjects were randomised. Baseline data is provided in Table 1.

Table 1
Subject baseline characteristics

Sex, n (%)	Male 17 (50)
Age, mean (range), years	44.2 (22–73)
DMFT index, mean (range)	12 (0–23)
Total Winkel score, mean (range)	4.3 (2–10)
Mouth odour history, n (%)	
Continuous mouth odour	12 (35.3)
Problems in morning and evening	10 (29.4)
Problems in morning only	12 (35.3)
Current use of products for mouth odour, n (%)	
Mouth rinse products	33 (97)
Chewing gum	23 (67.7)
Dental floss	23 (67.7)
Lozenges	19 (55.9)
	17 (50)
VSC concentration, ppb (SD)	
CB12/Water/Water sequence (n=18)	208.6 (100.0)
Water/CB12/CB12 sequence (n=16)	300.8 (346.1)

DMFT: decayed, missing and filled teeth; VSC: volatile sulphur compounds; ppb: parts per billion; SD: standard deviation

Efficacy

- CB12 produced a clear and durable effect (12 hour) on intra-oral halitosis compared to water, observed for all primary and secondary endpoints assessed (Table 2).
 - Use of CB12 was associated with a significant reduction in oral concentrations of hydrogen sulphide (Figure 1), methyl mercaptan (Figure 2), dimethyl sulphite and VSCs (halimetry) both overnight (12 hour after first rinse) and during the day (12 hour after the second rinse).
 - Patients' organoleptic score also significantly (P=0.0043) reduced overnight following CB12 use.

Table 2
Difference in *least square* mean change from baseline (by ANOVA/ANCOVA), across all three treatment periods (*P-values are one sided*)

	Difference in mean change from baseline			
	Overnight assessment (12 hour after the first rinse)		Daytime assessment (12 hour after the second rinse)	
	Diff (95% CI)	P-value	Diff (95% CI)	P-value
Hydrogen sulphide CB12-Water	-1.409 (-1.826, -0.993)	P<0.0001	-1.706 (-2.257, -1.155)	P<0.0001
Methyl mercaptan CB12-Water	-1.054 (-1.420; -0.688)	P<0.0001	-1.566 (-2.200, -0.933)	P<0.0001
Dimethyl sulphide CB12-Water	-0.757 (-1.158, -0.356)	P=0.0003	-0.849 (-1.335, -0.362)	P=0.0007
VSCs (halimetry) CB12-Water	-0.503 (-0.655, -0.352)	P<0.0001	-0.509 (-0.676, -0.342)	P<0.0001
Organoleptic score CB12-Water	-0.5 (-0.9, -0.1)	P=0.0043	-0.3 (-0.7, -0.1)	P=0.0458

Diff: difference; CI: confidence interval

Figure 1
Mean hydrogen sulphide over time (ITT population, aggregation of treatment periods)

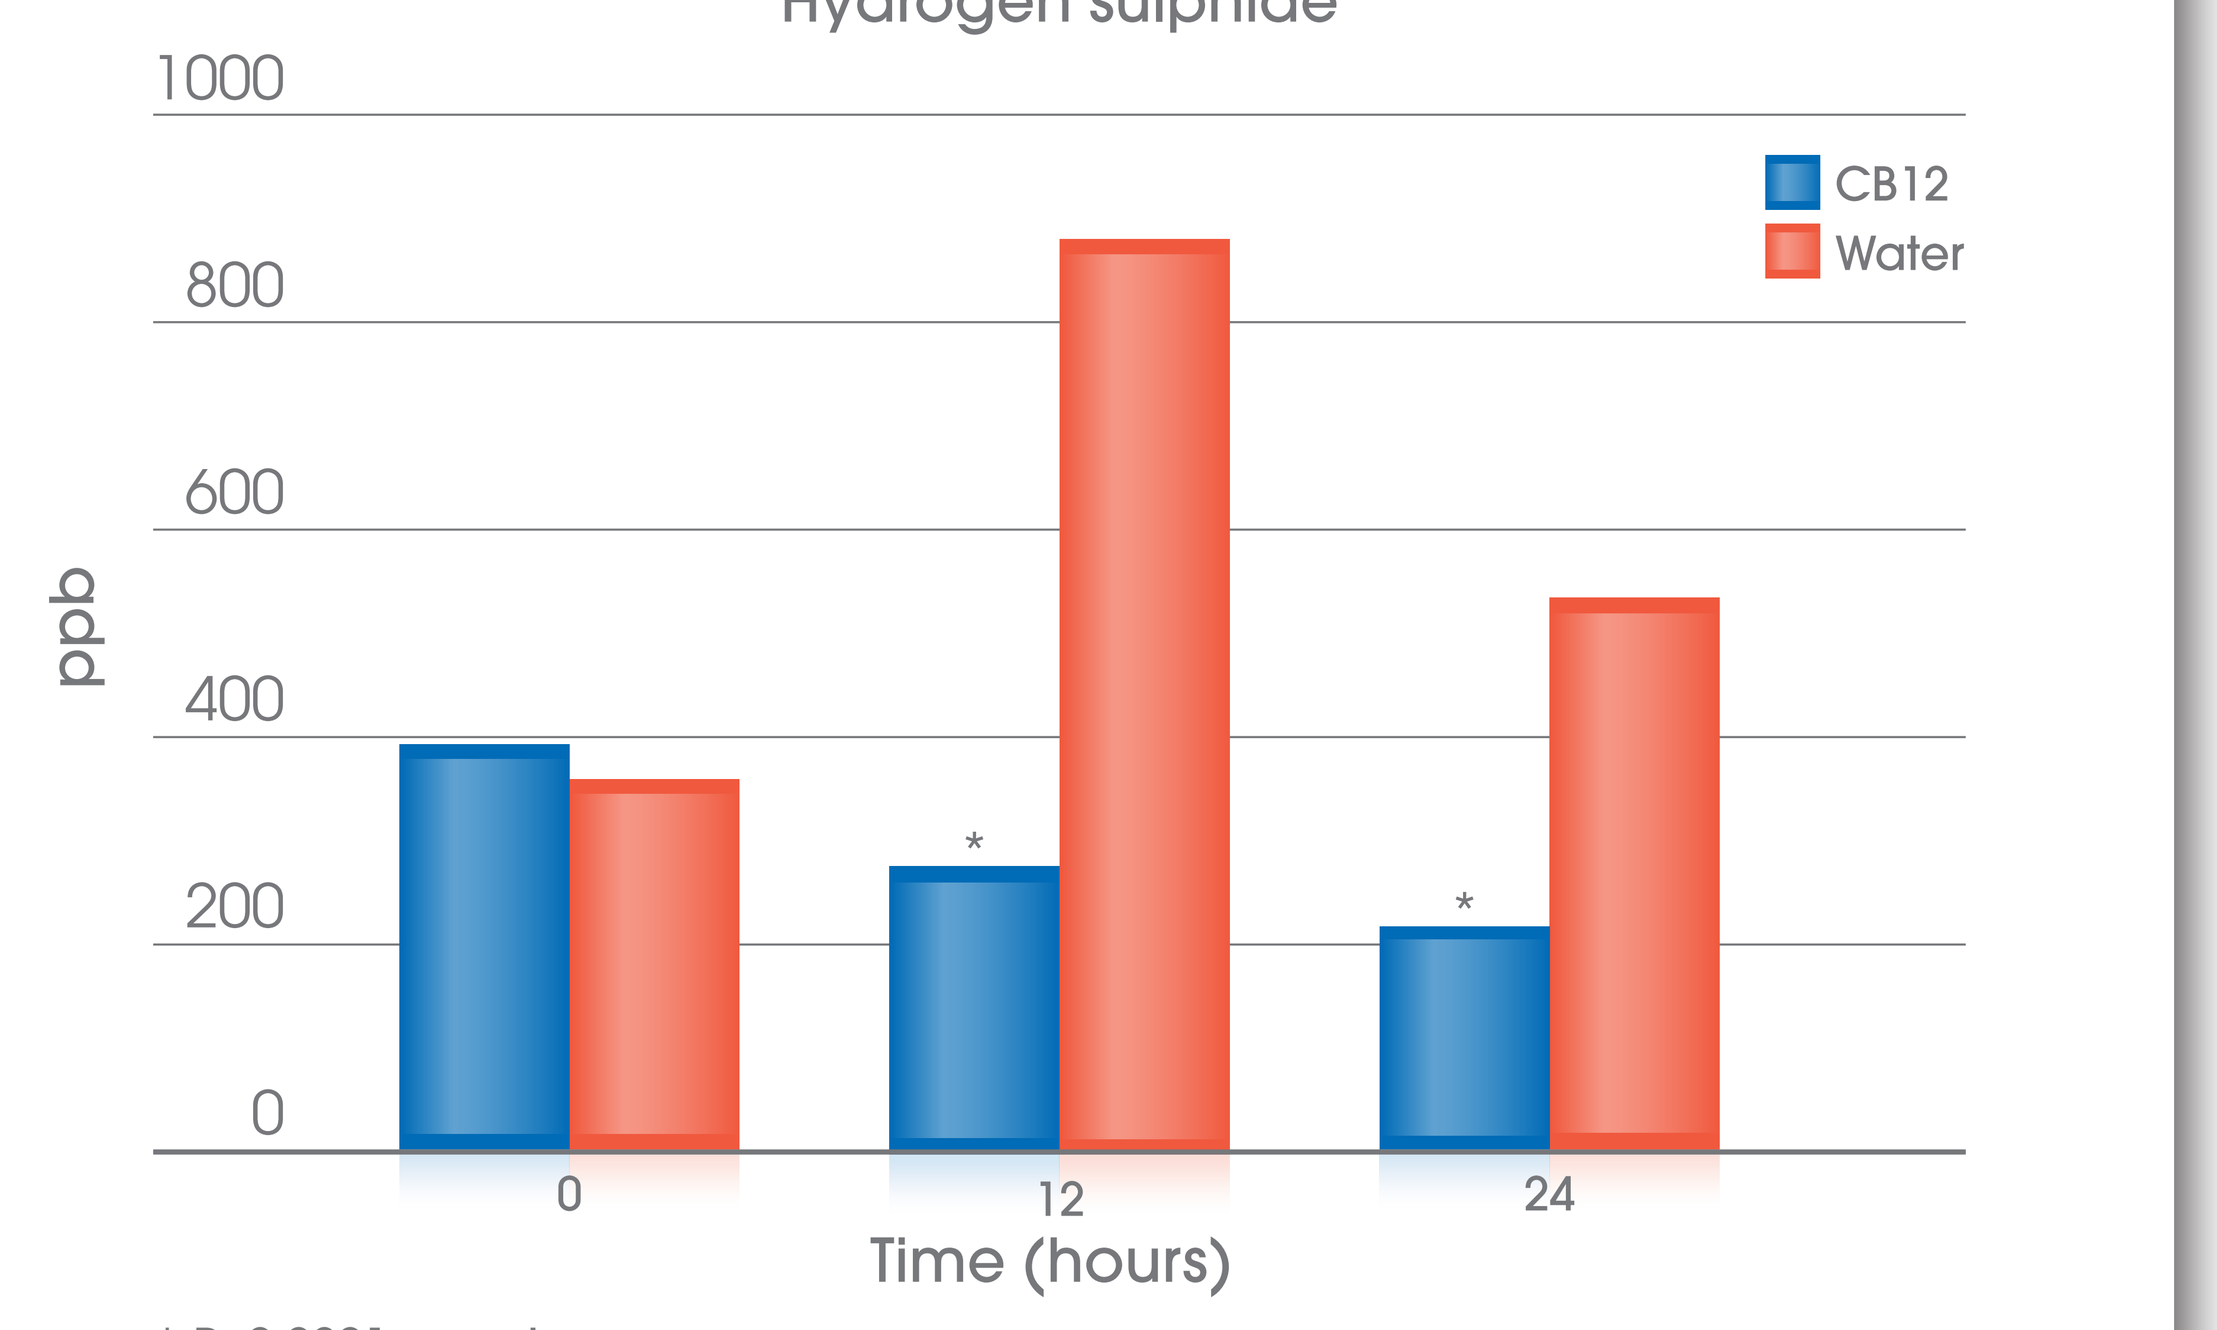
Hydrogen sulphide
1000
800
600
400
200
0
ppb
CB12
Water
0
12
24
Time (hours)
* P<0.0001 vs water

Figure 2
Mean methyl mercaptan over time (ITT population, aggregation of treatment periods)

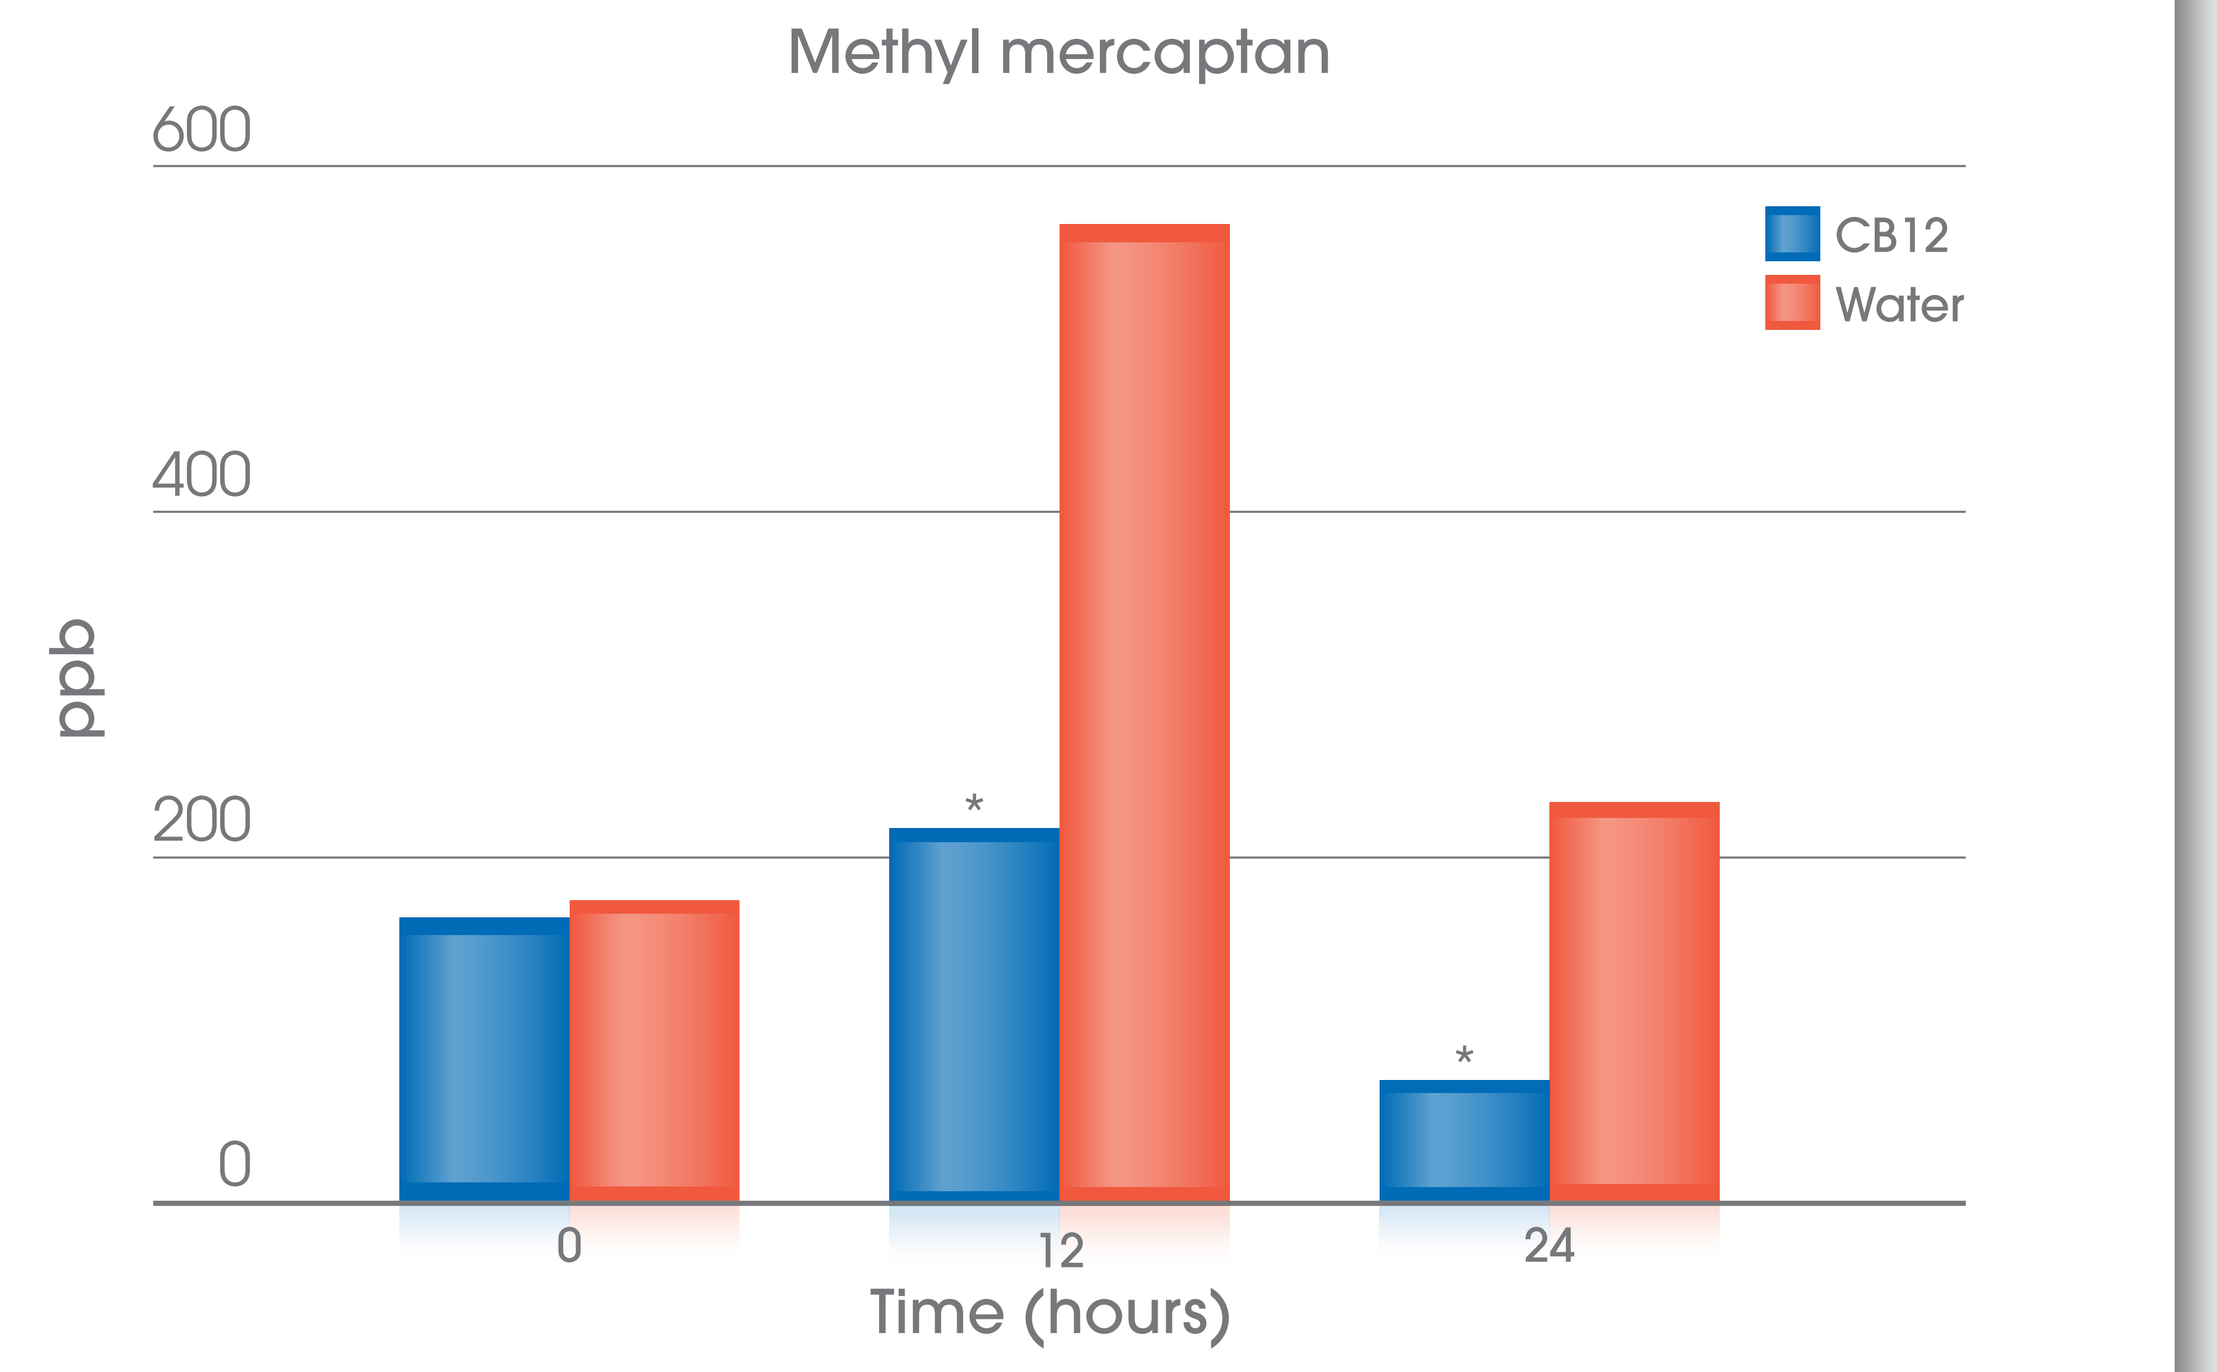
Methyl mercaptan
600
400
200
0
ppb
CB12
Water
0
12
24
Time (hours)
* P<0.0001 vs water

- Six subjects reported treatment-emergent adverse events (TEAE) following CB12 and five following water. Two TEAEs were considered related to CB12: application site reaction and dysgeusia (n=1 each; 2.9%). Both were classified as mild and were resolved.

Conclusions

- This well-designed, state-of-the-art study provides subjective and objective evidence that CB12 is an effective and long-lasting (at least 12 hours) treatment for halitosis, both overnight and during the day.
- These results confirm findings from other CB12 studies.
- So CB12 is backed by robust clinical data making it recommendable for evidence-based treatment of oral halitosis.

References

1. Liu XN, et al. J Clin Periodontol 2006; 33:31–6; **2.** Outhouse TL, et al. Cochrane Database Syst Rev 2006;2:CD005519; **3.** Porter SR, Scully C. BMJ 2006; 333:632–5; **4.** Bornstein MM, et al. Eur J Oral Sci 2009; 117:261–7; **5.** Tonzetich J. J Periodontol 1977; 48:13–20; **6.** Seemann R, et al. J Breath Res 2014; 8:017101; **7.** Greenman J, et al. J Breath Res 2014; 8:017102; **8.** Tangerman A, Winkel EG. J Breath Res 2008; 2:017010; **9.** Vandekerckhove B, et al. J Clin Periodontol 2009; 36:964-9; **10.** Thrane P, et al. Dental Health 2009;48:2–6; **11.** Saad S, et al. Oral Dis 2011; 17:180–6; **12.** Greenman J, et al. J Dent Res 2004; 83:81–5.

Funding: This study was funded by Meda. Acknowledgments: Medical writing assistance was provided by Medscript Ltd, funded by Meda. Conflict of interest statements: All authors have no conflict of interest to report

Scan here to download this poster and others in the series